Treatment for Youth Experiencing Harms from Substance Use across the Continuum of Care

Jill Fairbank, Sheena Taha and Sarah Wallingford | Canadian Centre on Substance Use and Addiction

Why Youth?

- 15–25 year old Canadians use substances more than adults
- Substance use can harm brain development occurring during youth

Response

CCSA combined research and experiential evidence to outline targeted treatment for youth and principles of practice

Collaboration

The continuum includes input from:

- Addictions workers
- Family physicians
- Pain specialists
- Pediatricians
- Psychiatrists
- Psychologists
- Researchers
- School counsellors
- Youth

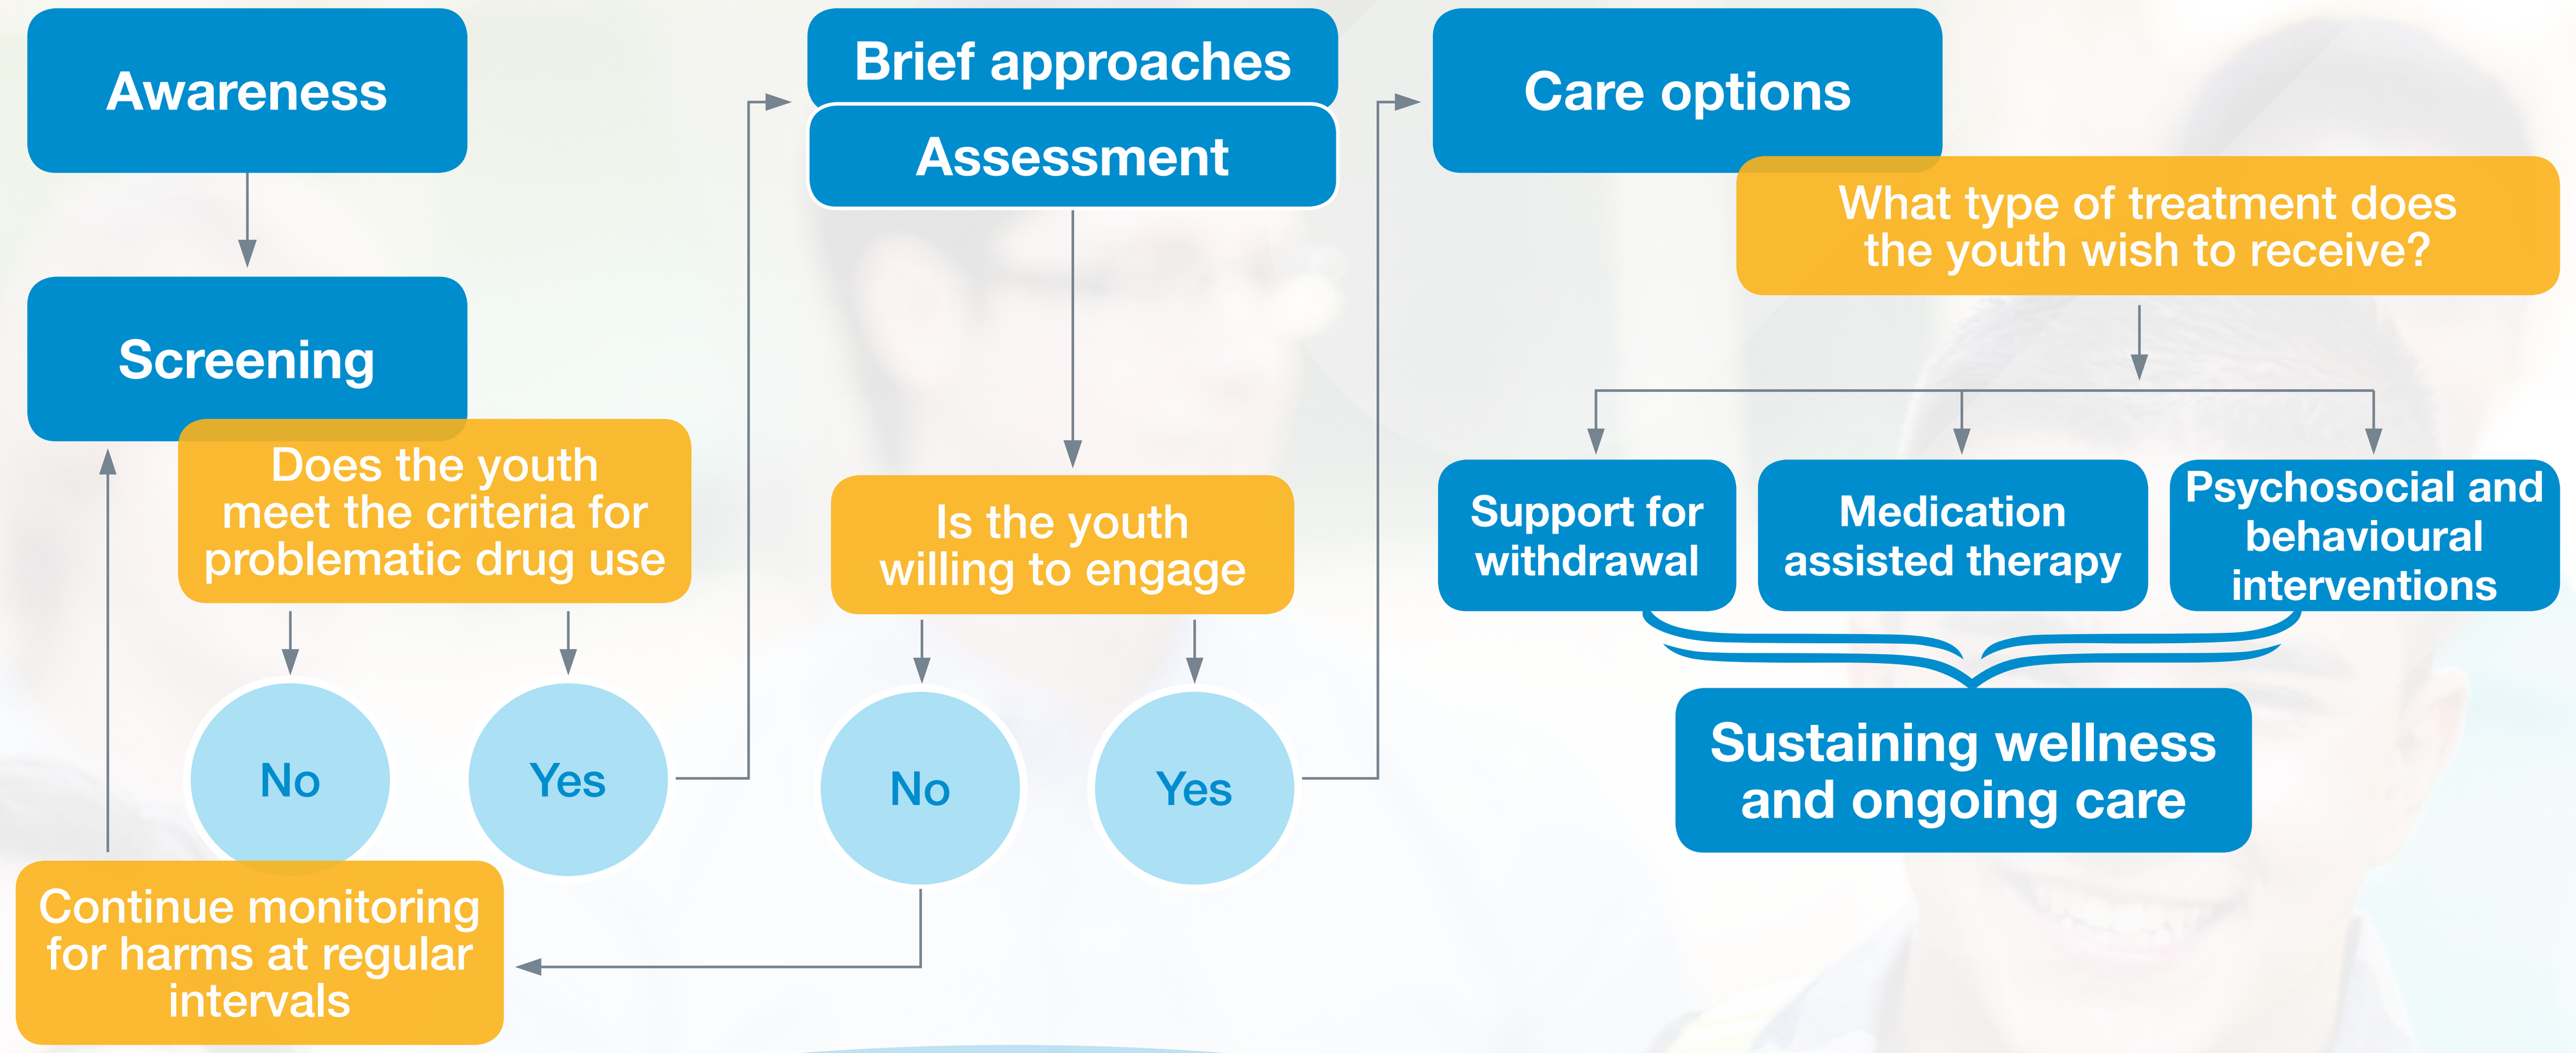
Awareness
Screening
Does the youth meet the criteria for problematic drug use
No
Yes
Continue monitoring for harms at regular intervals
Brief approaches
Assessment
Is the youth willing to engage
No
Yes
Care options
What type of treatment does the youth wish to receive?
Support for withdrawal
Medication assisted therapy
Psychosocial and behavioural interventions
Sustaining wellness and ongoing care

Guiding Principles

The use of tools and services available vary by jurisdiction, so guiding principles:

- Provide considerations that apply to all care
- Are central to a youth-friendly approach
- Should be applied across the continuum of care

Principle 1

Develop and Maintain a Trusting Relationship

- Effective screening occurs after rapport is developed
- Have open door policy to ensure wellness journey

Principle 2

Care Is Patient Centred and Client Centred

- Allow youth to lead conversation
- Develop an individualized care plan that can be adjusted over time

Principle 3

Use Integrated Approach to Address All Concerns

- Consider stage of change, age and development phase, culture and context of care
- Ensure mental and physical health are also addressed
- Use trauma- and gender-informed approaches

Principle 4

Focus on Protective Factors and Building Resiliency

- Use a strength-based approach

Principle 5

Embed Harm Reduction across the Continuum

- Apply health promotion and prevention principles
- Consider youth's need for information about safe use of substances and safe spaces

Outcome

Treatment is a non-linear process that might require the individual to experiment with a multitude of pathways to determine which approach — or combination of approaches — is most suitable for them

A de-stigmatizing, supportive environment that meets individuals' needs will provide the best opportunity for success

Canadian Centre on Substance Use and Addiction
Evidence. Engagement. Impact.

Contact Information
Sheena Taha, PhD
Knowledge Broker, CCSA
613-235-4048 ext. 229 or staha@ccsa.ca
75 Albert Street, Suite 500
Ottawa ON K1P 5E7, Canada

Production of this poster has been made possible through a financial contribution from Health Canada.
The views expressed herein do not necessarily represent the views of Health Canada.
© Canadian Centre on Substance Use and Addiction 2017

Traitement de jeunes subissant les méfaits de la consommation de substances tout au long du continuum de soins

Jill Fairbank, Sheena Taha et Sarah Wallingford | Centre canadien sur les dépendances et l'usage de substances

Pourquoi les jeunes?

- Les jeunes Canadiens de 15 à 25 ans consomment plus que les adultes
- La consommation de substances peut nuire au développement du cerveau qui a lieu pendant la jeunesse

Réponse

Le CCDUS a combiné la recherche et les données expérientielles pour tracer les grandes lignes des traitements ciblés pour jeunes et des principes de pratique

Collaboration

Le continuum comprend la contribution de :

- Travailleurs en dépendance
- Médecins de famille
- Spécialistes de la douleur
- Pédiatres
- Psychiatres
- Psychologues
- Chercheurs
- Conseillers scolaires
- Jeunes

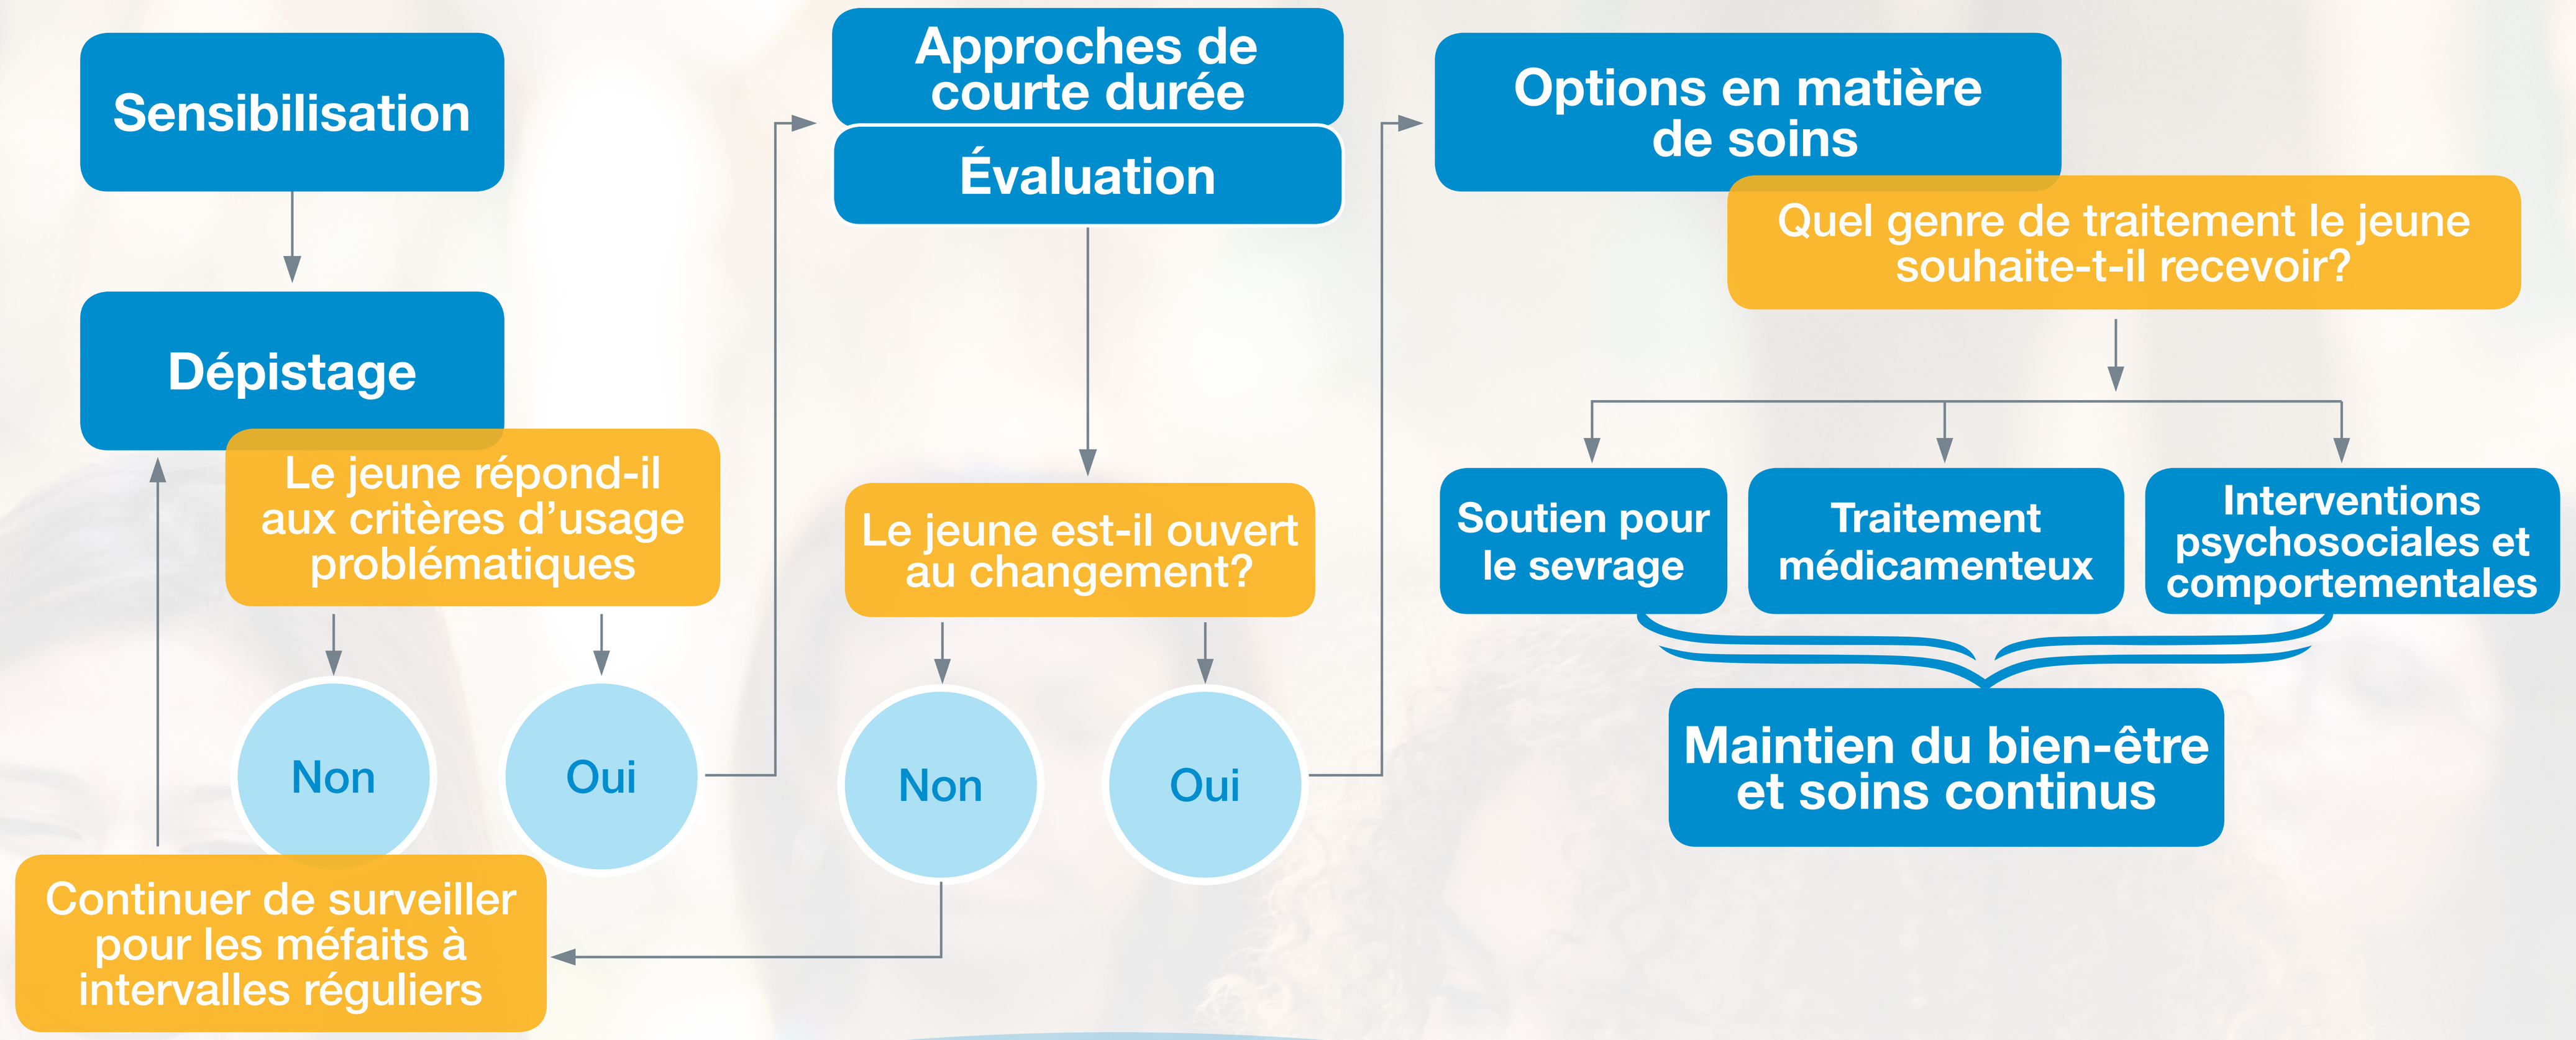
Sensibilisation
Dépistage
Le jeune répond-il aux critères d'usage problématiques
Non
Oui
Continuer de surveiller pour les méfaits à intervalles réguliers
Approches de courte durée
Évaluation
Le jeune est-il ouvert au changement?
Non
Oui
Options en matière de soins
Quel genre de traitement le jeune souhaite-t-il recevoir?
Soutien pour le sevrage
Traitement médicamenteux
Interventions psychosociales et comportementales
Maintien du bien-être et soins continus

Principes directeurs

L'utilisation des outils et services offerts varie par compétence. Par conséquent, les principes directeurs :

- proposent des considérations s'appliquant à tous les soins prodigués
- sont essentiels à une approche convenant aux jeunes
- doivent être appliqués tout au long du continuum de soins

Principe 1

Établir et entretenir une relation de confiance

- Établir un solide rapport pour assurer un dépistage efficace
- Adopter une politique de porte ouverte pour assurer un cheminement de bien-être

Principe 2

Axer les soins vers le patient et permettre au client de les diriger

- Permettre au jeune d'orienter la conversation
- Dresser un plan de soins personnalisé pouvant être modifié au fil du temps

Principe 3

Utiliser une approche intégrée pour aborder les préoccupations

- Tenir compte de l'étape du changement, de l'âge et de l'étape de développement, de la culture et du contexte de soins
- Aborder aussi la santé mentale et physique
- Adopter des approches sensibles au traumatisme et au genre

Principe 4

Mettre l'accent sur les facteurs de protection et le renforcement de la résilience

- Adopter une approche axée sur les forces

Principe 5

Incorporer la réduction des méfaits à l'ensemble du continuum

- Appliquer les principes de la promotion de la santé et de la prévention des méfaits
- Tenir compte des besoins d'information des jeunes sur l'usage sécuritaire des substances et les espaces sécuritaires

Résultats

Le traitement est un processus non linéaire qui pourrait exiger que la personne fasse l'essai de nombreux protocoles pour déterminer lequel ou lesquels lui conviennent le mieux

Un milieu déstigmatisant et bienveillant qui répond aux besoins des particuliers offrira la meilleure chance de réussite.

Centre canadien sur les dépendances et l'usage de substances
Données. Engagement. Résultats.

Contacter :
Sheena Taha, Ph.D.
Courtière du savoir, CCDUS
613-235-4048, poste 229 ou staha@ccsa.ca
500–75 rue Albert
Ottawa ON K1P 5E7, Canada

Cette affiche a été produite grâce à la contribution financière de Santé Canada.
Les opinions exprimées ne représentent pas nécessairement celles de Santé Canada.
© Centre canadien sur les dépendances et l'usage de substances, 2017